Development and validation of a PBPK model of gepotidacin in rats to predict exposure in human prostate

Vincent Aranzana-Climent, Valentin Al Jalali, Hwa-Ping Feng, Karen O'Dwyer, William Couet, Markus Zeitlinger, Sandrine Marchand

▶ **To cite this version:**

Vincent Aranzana-Climent, Valentin Al Jalali, Hwa-Ping Feng, Karen O'Dwyer, William Couet, et al.. Development and validation of a PBPK model of gepotidacin in rats to predict exposure in human prostate. 34th European Congress of Clinical Microbiology and Infectious Diseases (ECCMID), Apr 2024, Barcelona, Spain. hal-04571063

HAL Id: hal-04571063
https://hal.science/hal-04571063

Submitted on 7 May 2024

HAL is a multi-disciplinary open access archive for the deposit and dissemination of scientific research documents, whether they are published or not. The documents may come from teaching and research institutions in France or abroad, or from public or private research centers.

L'archive ouverte pluridisciplinaire **HAL**, est destinée au dépôt et à la diffusion de documents scientifiques de niveau recherche, publiés ou non, émanant des établissements d'enseignement et de recherche français ou étrangers, des laboratoires publics ou privés.

P2492

Development and validation of a PBPK model of gepotidacin in rats to predict exposure in human prostate

Vincent Aranzana-Climent[1], Valentin Al Jalali[2], Hwa-Ping Feng[3], Karen O'Dwyer[3], William Couet[1,4], Markus Zeitlinger[2], Sandrine Marchand[1,4]

Contact: vincent.aranzana.climent@univ-poitiers.fr

[1] Université de Poitiers, INSERM U1070, France [2] Medical University of Vienna, Austria [3] GSK, Collegeville, United States [4] CHU de Poitiers, France

Introduction

Gepotidacin is a novel antibiotic currently under phase 3 development for uncomplicated urinary tract infection and uncomplicated gonorrhea. The objective of the current study was to assess the possibility of predicting its human prostate pharmacokinetics by combining preclinical data and physiologically based pharmacokinetic (PBPK) modeling.

Materials and methods

1

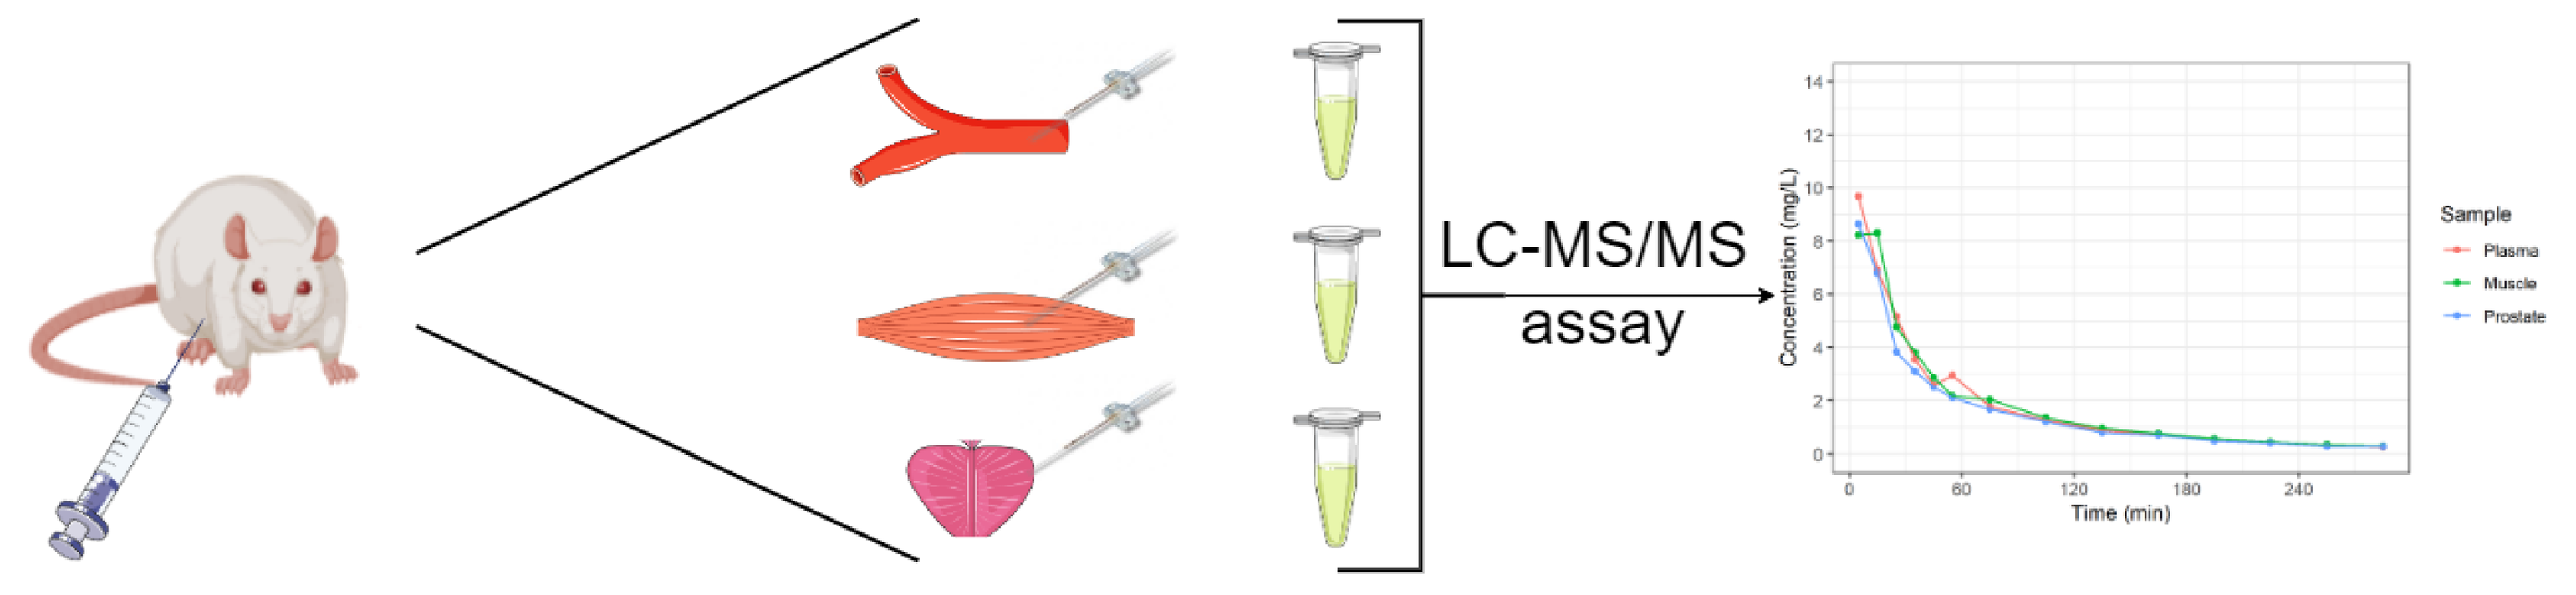
LC-MS/MS
assay
Sample
Plasma
Muscle
Prostate
Time (min)

20 rats Gepotidacin IV Bolus 20 mg/kg single dose

In vivo microdialysis serial sampling over 5h from blood, muscle and prostate

Unbound plasma, muscle and prostate gepotidacin concentrations

Step 1: PK study of gepotidacin pharmacokinetics in blood, muscle and prostate using multiple time-points *in vivo* microdialysis

2

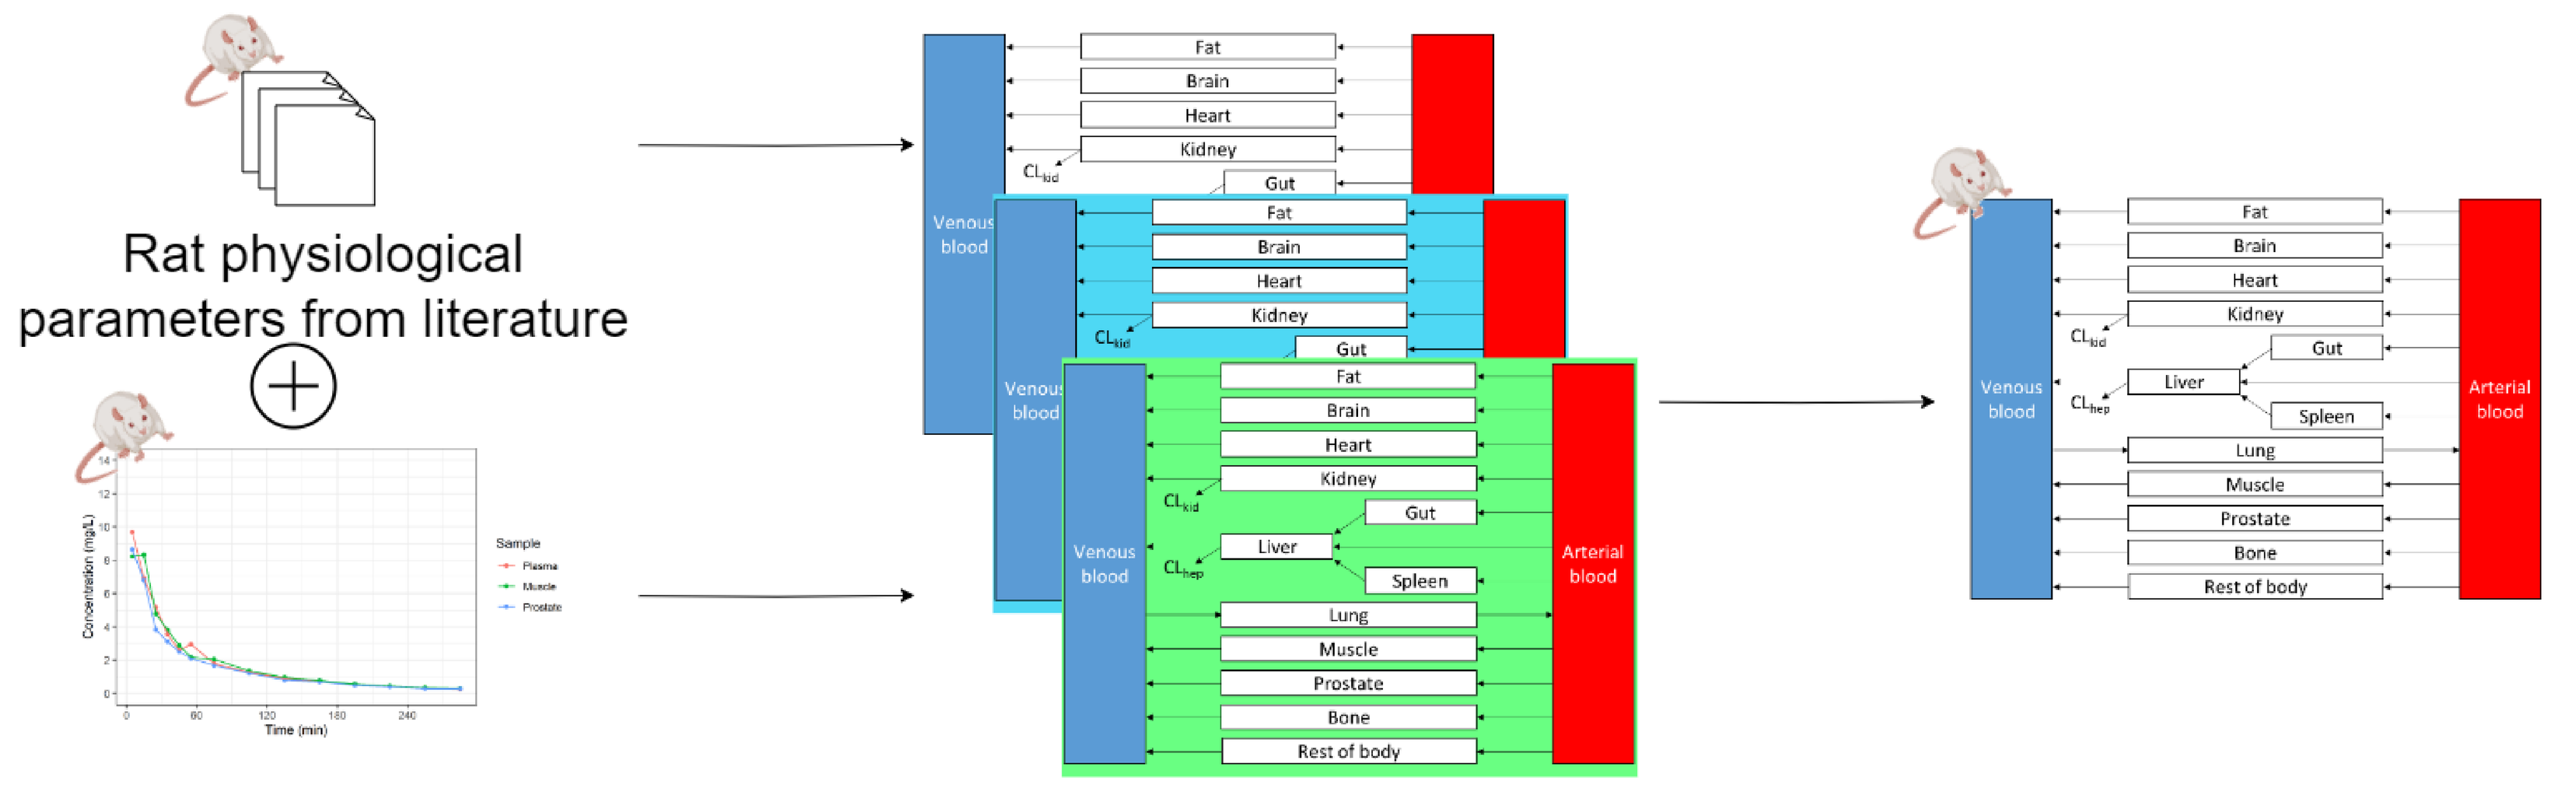
Rat physiological parameters from literature
Venous blood
Arterial blood
Fat
Brain
Heart
Kidney
Gut
Liver
Spleen
Lung
Muscle
Prostate
Bone
Rest of body

Rat unbound plasma, muscle and prostate data

PBPK model fitting and selection

Best rat PBPK model

Step 2: Development of a PBPK model for gepotidacin in rats

3

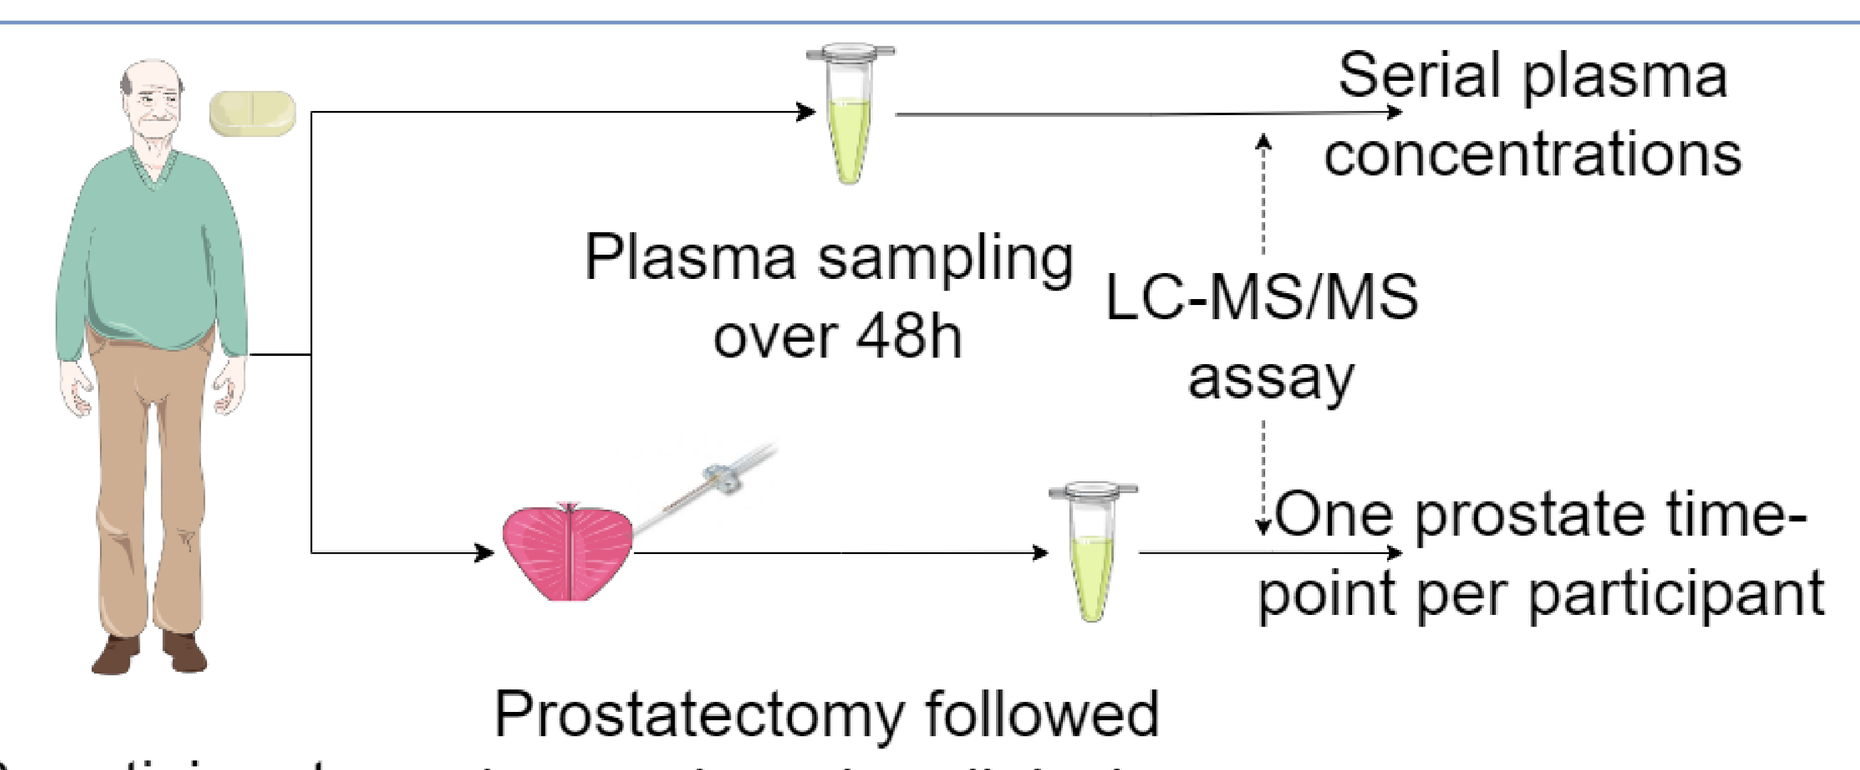
Serial plasma concentrations
Plasma sampling over 48h
LC-MS/MS assay
One prostate time-point per participant
Prostatectomy followed by *ex vivo* microdialysis

26 participants Oral Gepotidacin 1500 mg single dose

Step 3: Composite human prostate concentration-time profile constructed from ex vivo microdialysis samples in a clinical study (NCT04484740/P2463).

Animal ethics :
The project was authorized by the French Ministry of National Education and Research in accordance with the European directive on animal experimentation under n°23655_2020011711481557 in February 2020. The experiment took place in the animal care facilities of the University of Poitiers: PREBIOS.

4

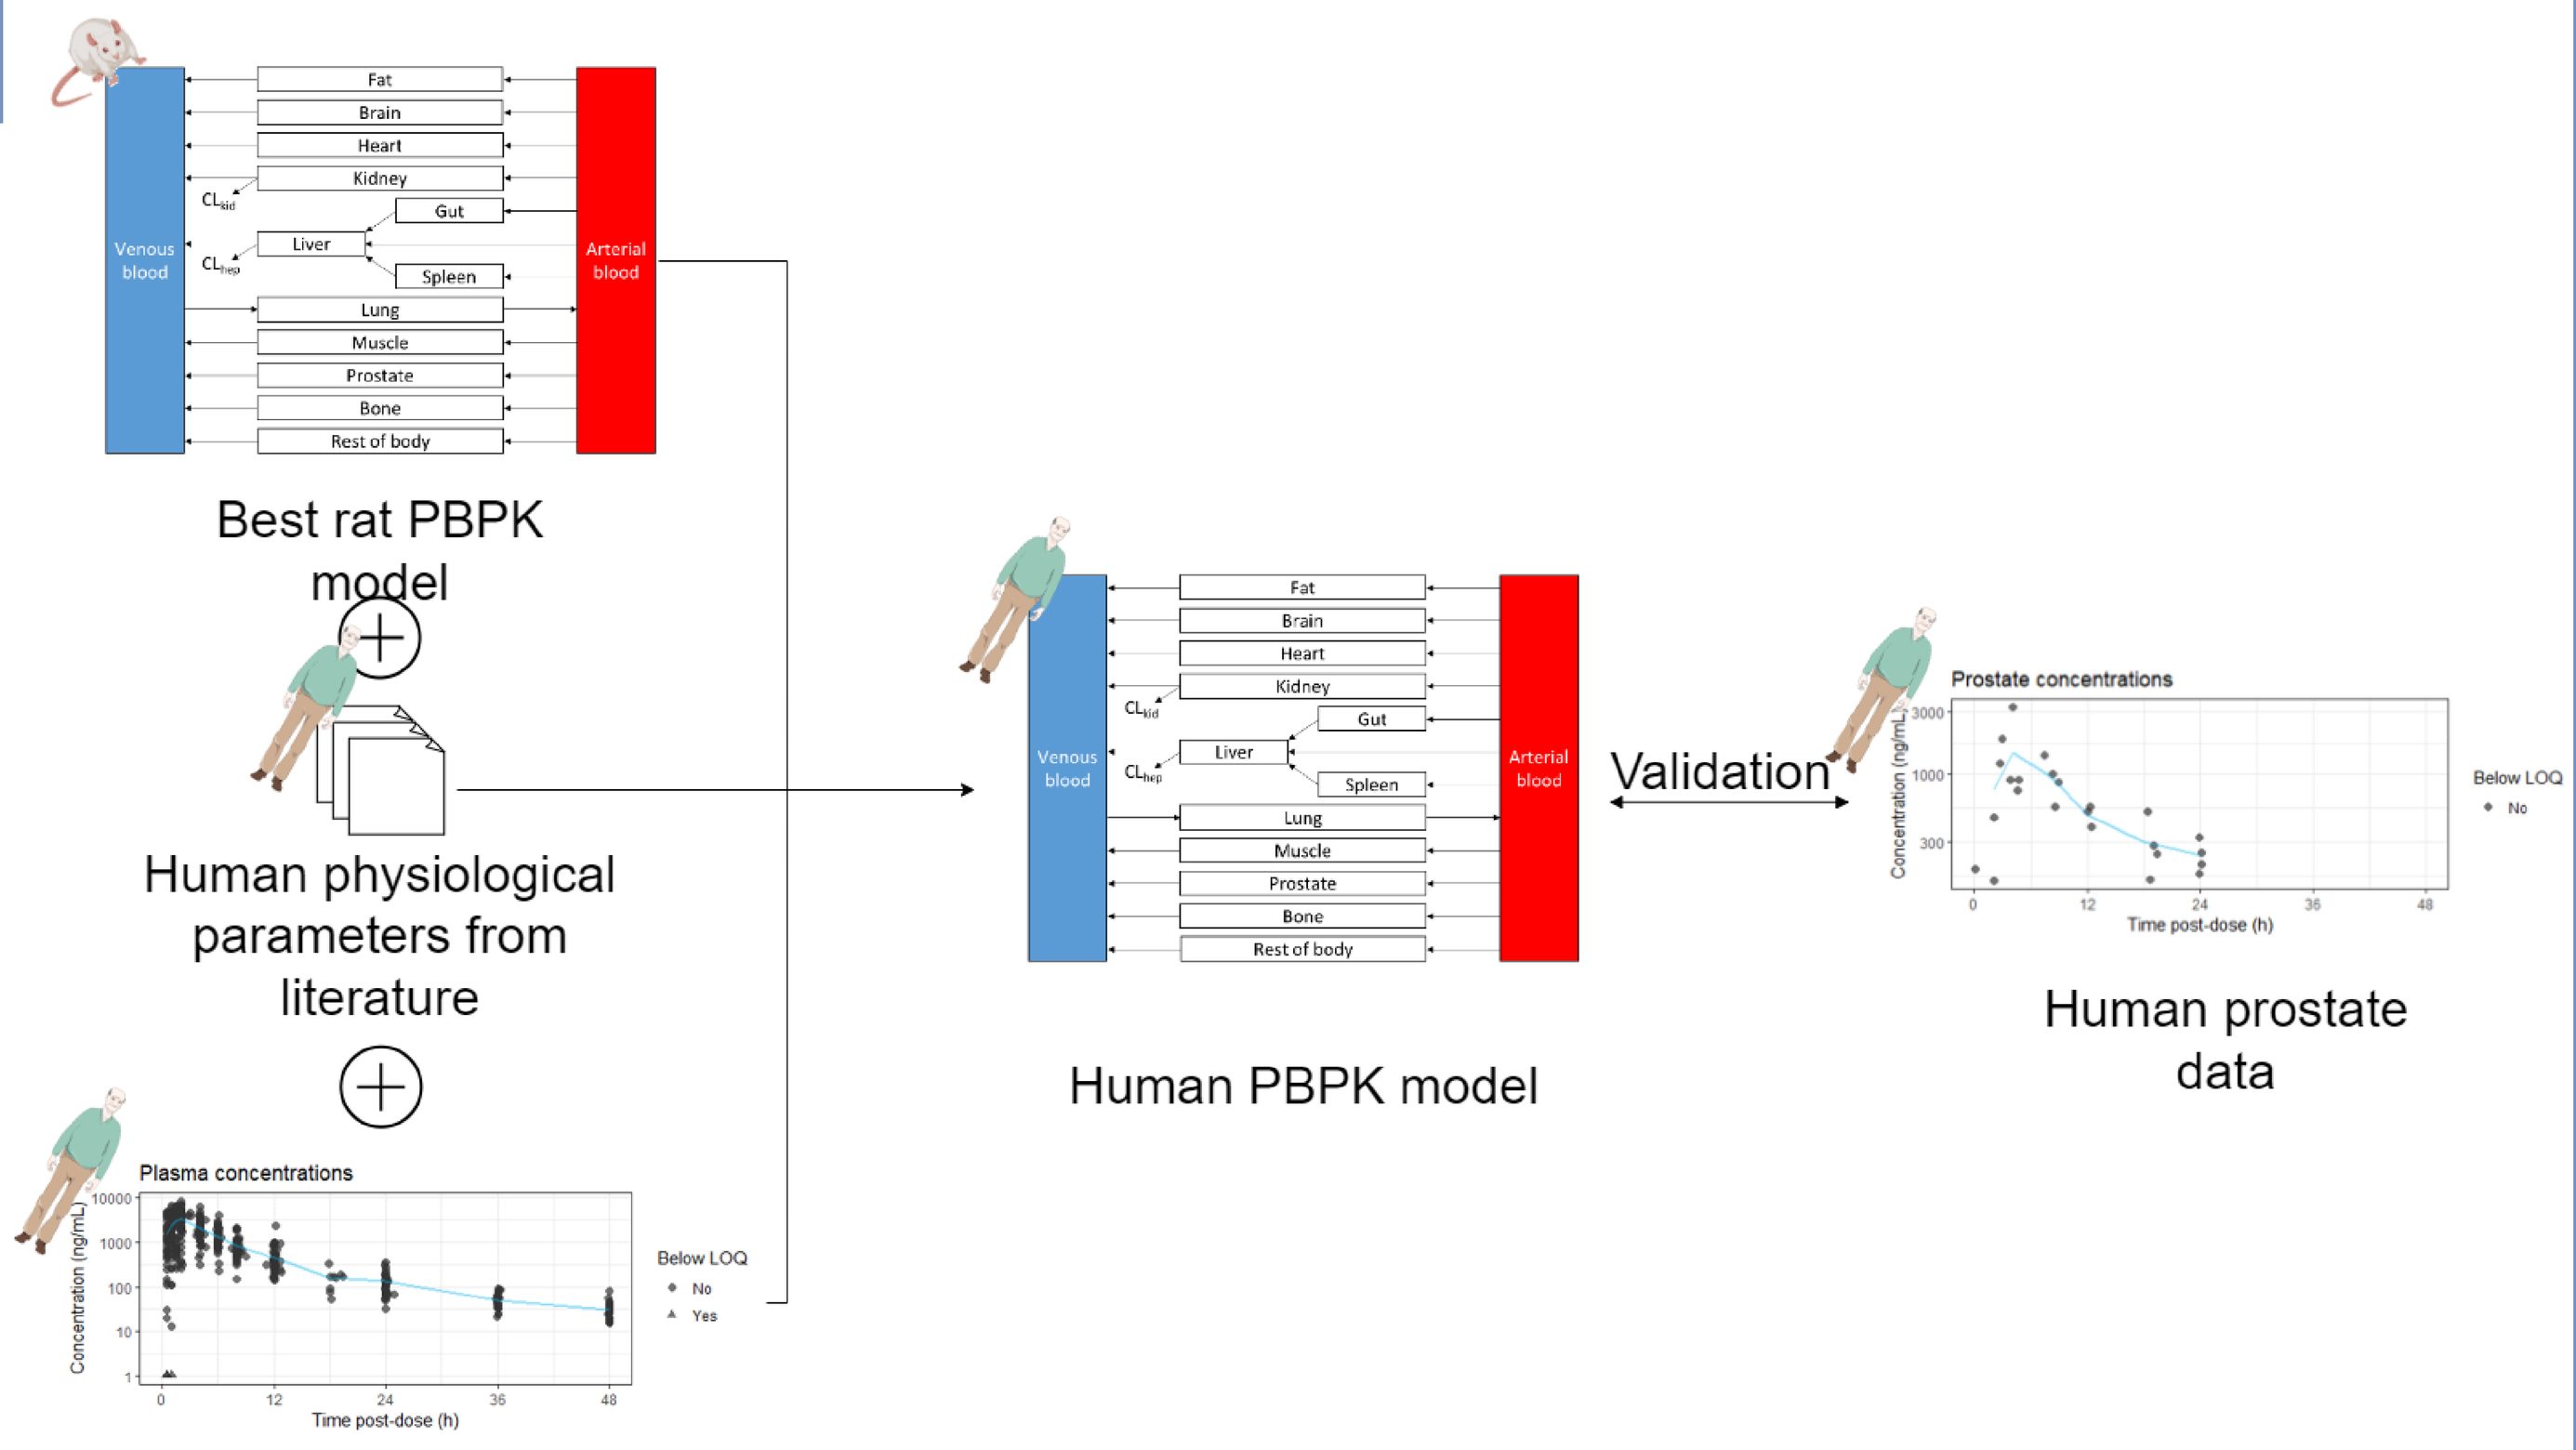
Best rat PBPK model
Human physiological parameters from literature
Plasma concentrations
Human plasma data
Human PBPK model
Validation
Prostate concentrations
Human prostate data

Step 4: Development and validation of a PBPK model for gepotidacin in humans

Mathematical methods and software :

- Three model structures were tested : Full PBPK, Reduced PBPK and PermQ PBPK (1)
- Three partition coefficient (Kp) calculation methods were tested, Rodgers and Rowland's traditional method (2), Rodgers and Rowland's method taking lysosomal trapping into account (3) and Schmitt's method (4)
- Rat prostate partition coefficients were determined experimentally, human prostate partition coefficient were fixed to the rat value
- Plasma PK parameter estimation was performed with Monolix (5).

Results

The best PBPK model (**Figure 1**) was a flow limited reduced PBPK model (Fig. 1a). Muscle and prostate were separated into vascular, interstitial and cellular spaces (Fig. 1b).

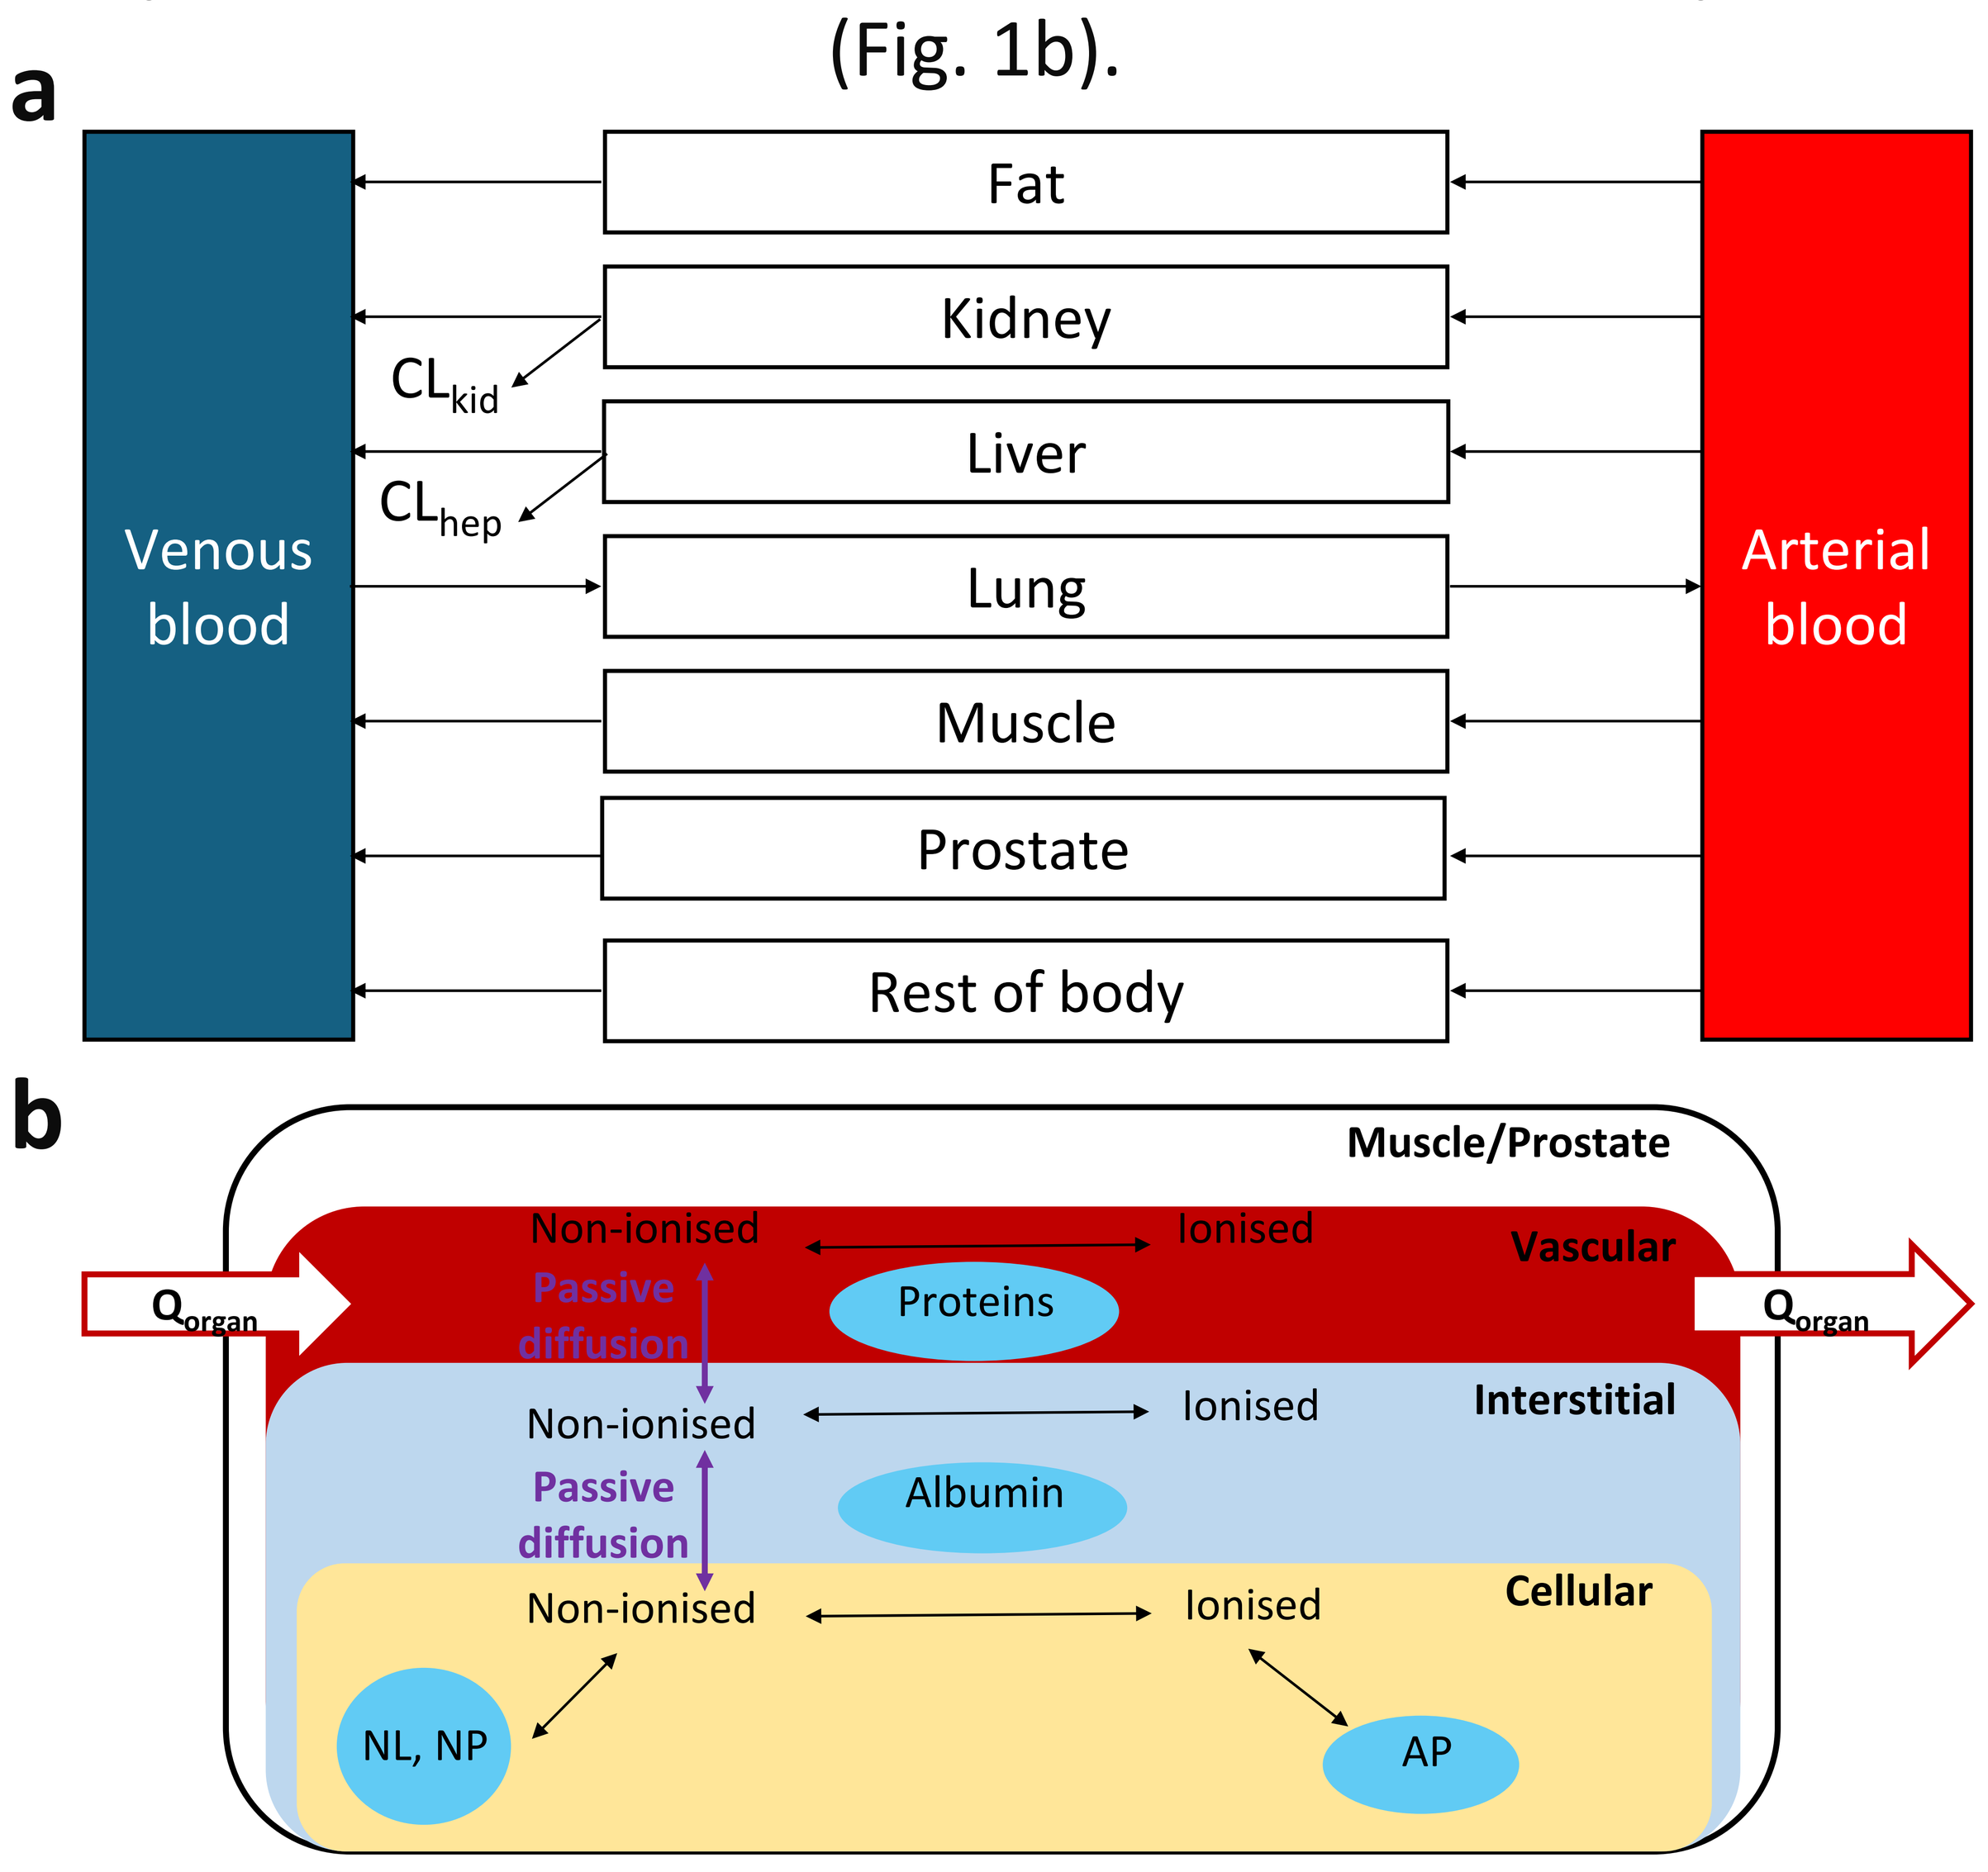
a
Venous blood
Fat
Kidney
Liver
Lung
Muscle
Prostate
Rest of body
Arterial blood
CL_{kid}
CL_{hep}
b
Muscle/Prostate
Non-ionised
Ionised
Vascular
Passive diffusion
Proteins
Q_{organ}
Interstitial
Albumin
Cellular
NL, NP
AP

Figure 1: Best PBPK model structure

As seen on **Figure 2**, rat unbound plasma, muscle and prostate concentrations were adequately described by the PBPK model with more than 80% of the predicted concentrations within 2-fold of the observed concentrations.

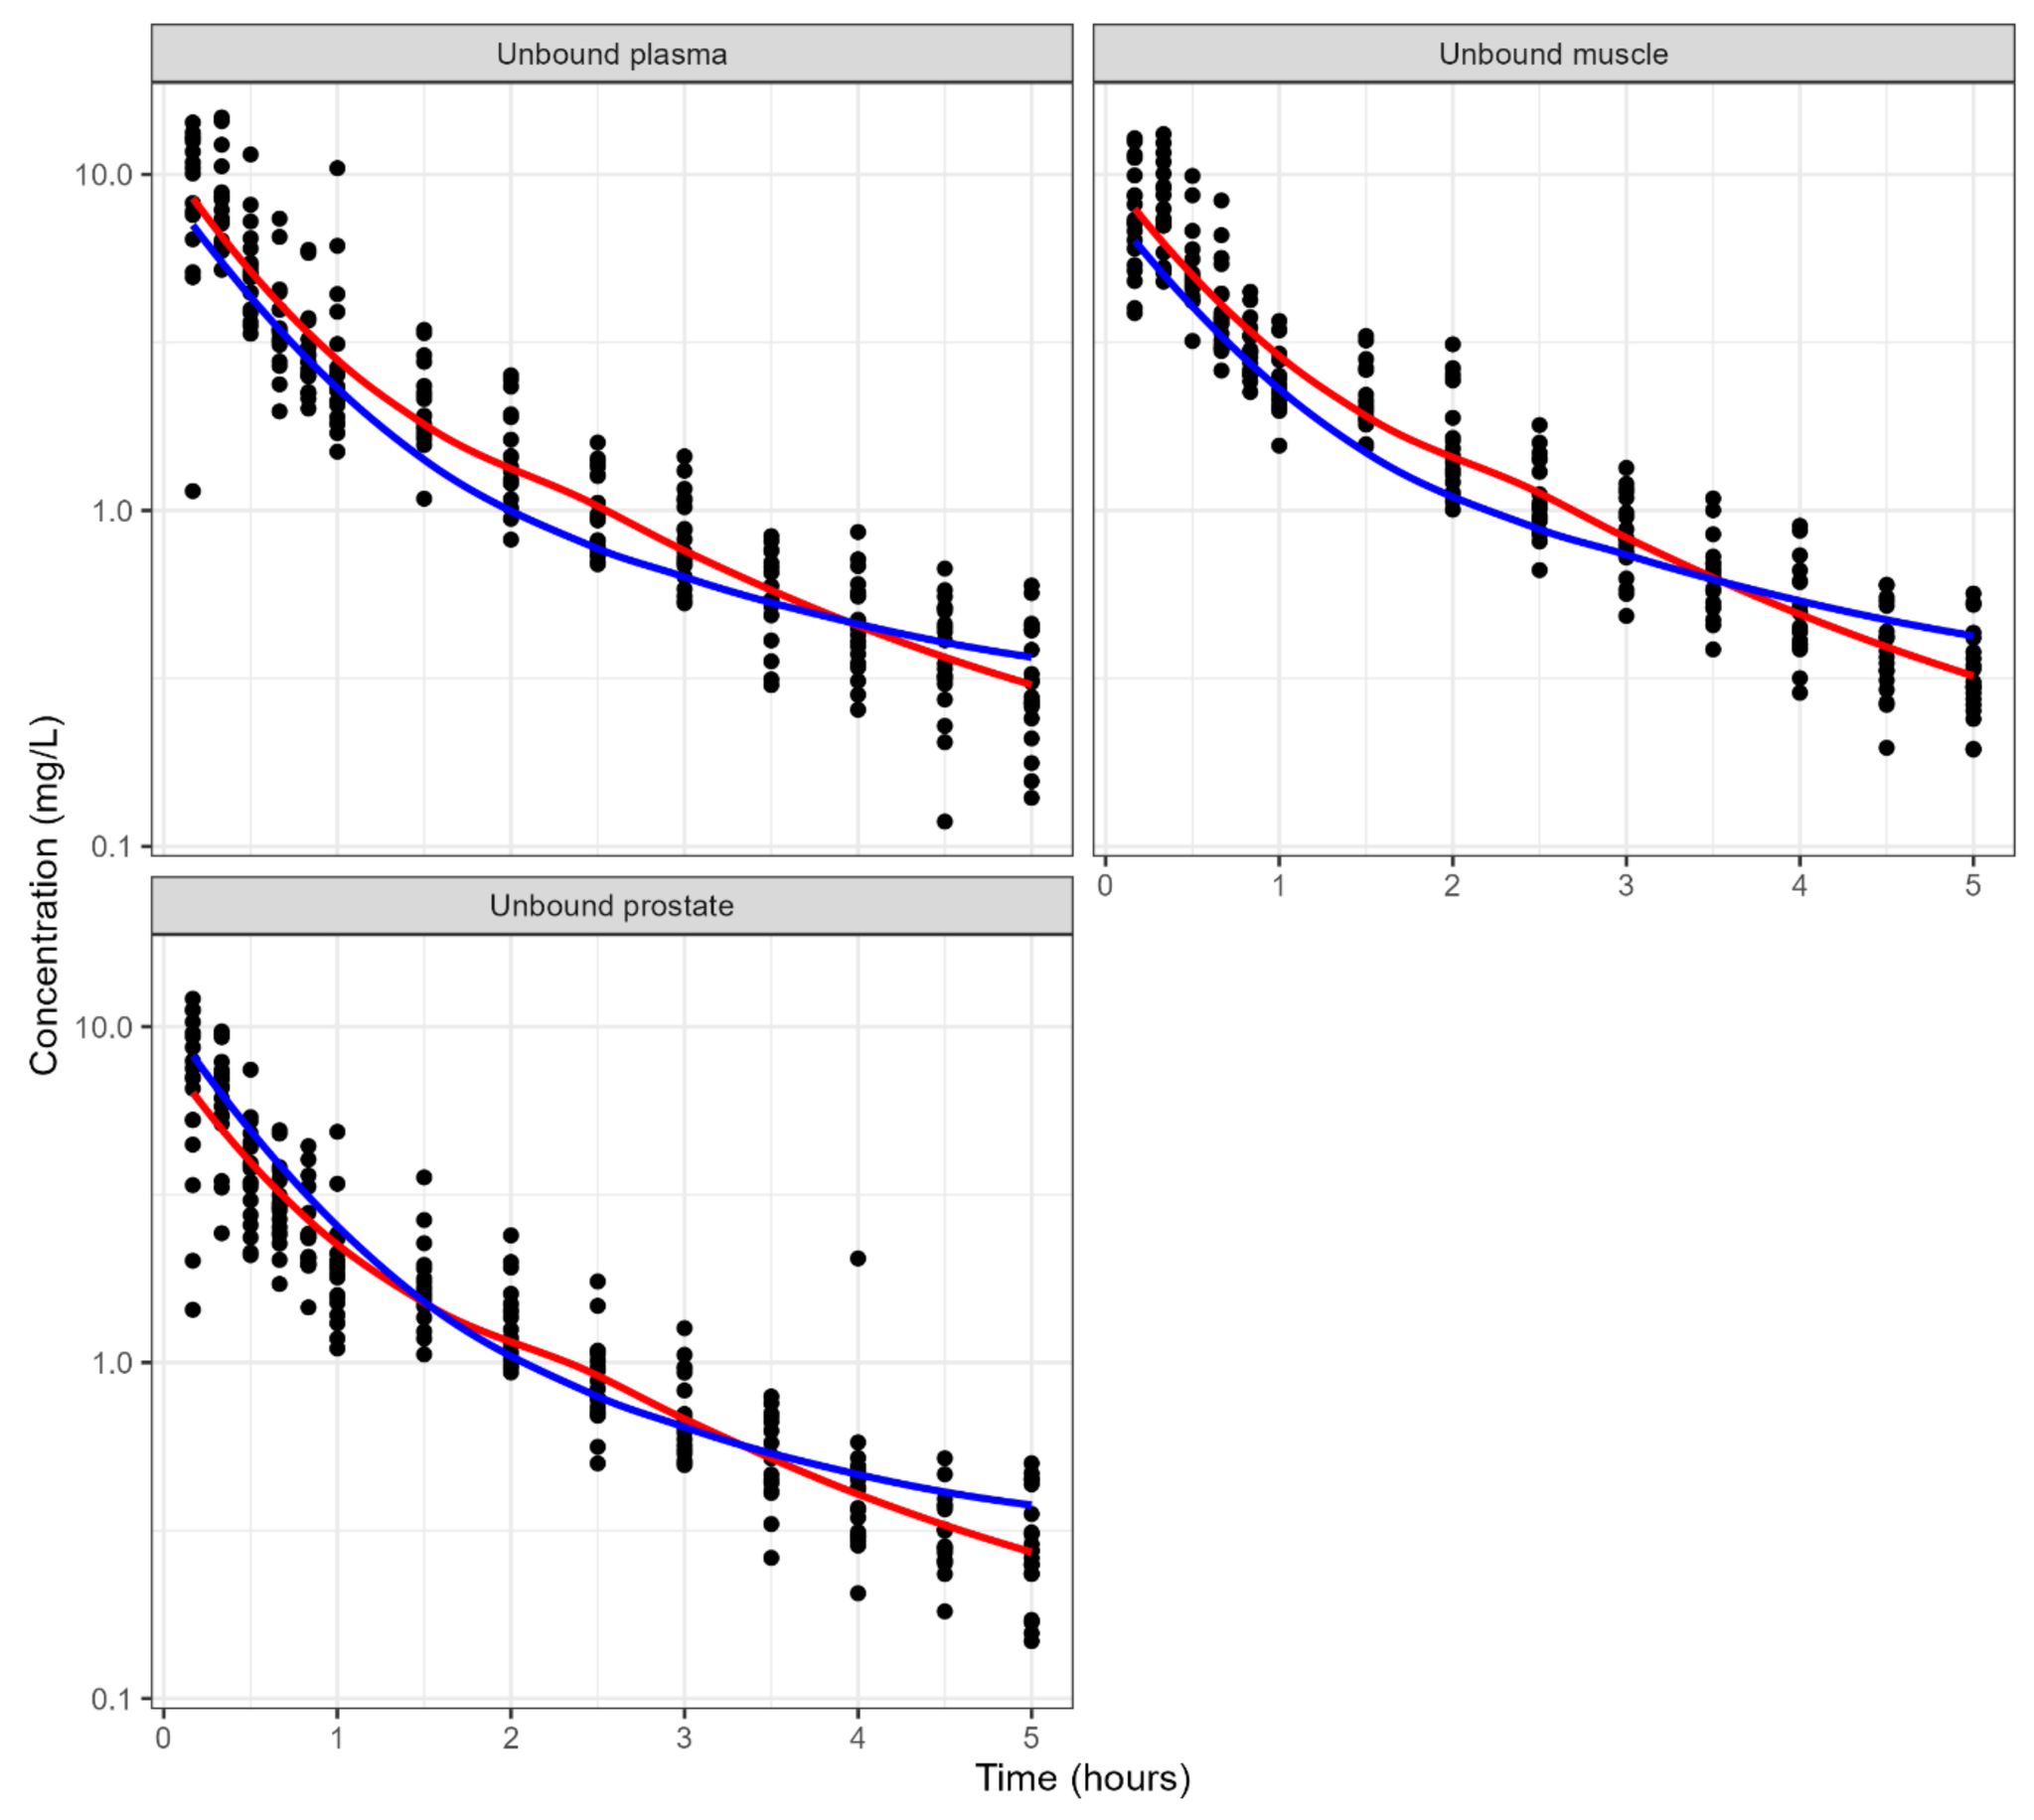
Unbound plasma
Unbound muscle
Unbound prostate
Concentration (mg/L)
Time (hours)

Figure 2: Rat measured concentrations (points) with observation mean (red) and prediction mean (blue).

As seen on **Figure 3**, there is a general agreement between the mean predicted prostate concentrations and the observed mean unbound prostate concentrations with 66.7% of the of the predicted concentrations within 2-fold of the observed concentrations.

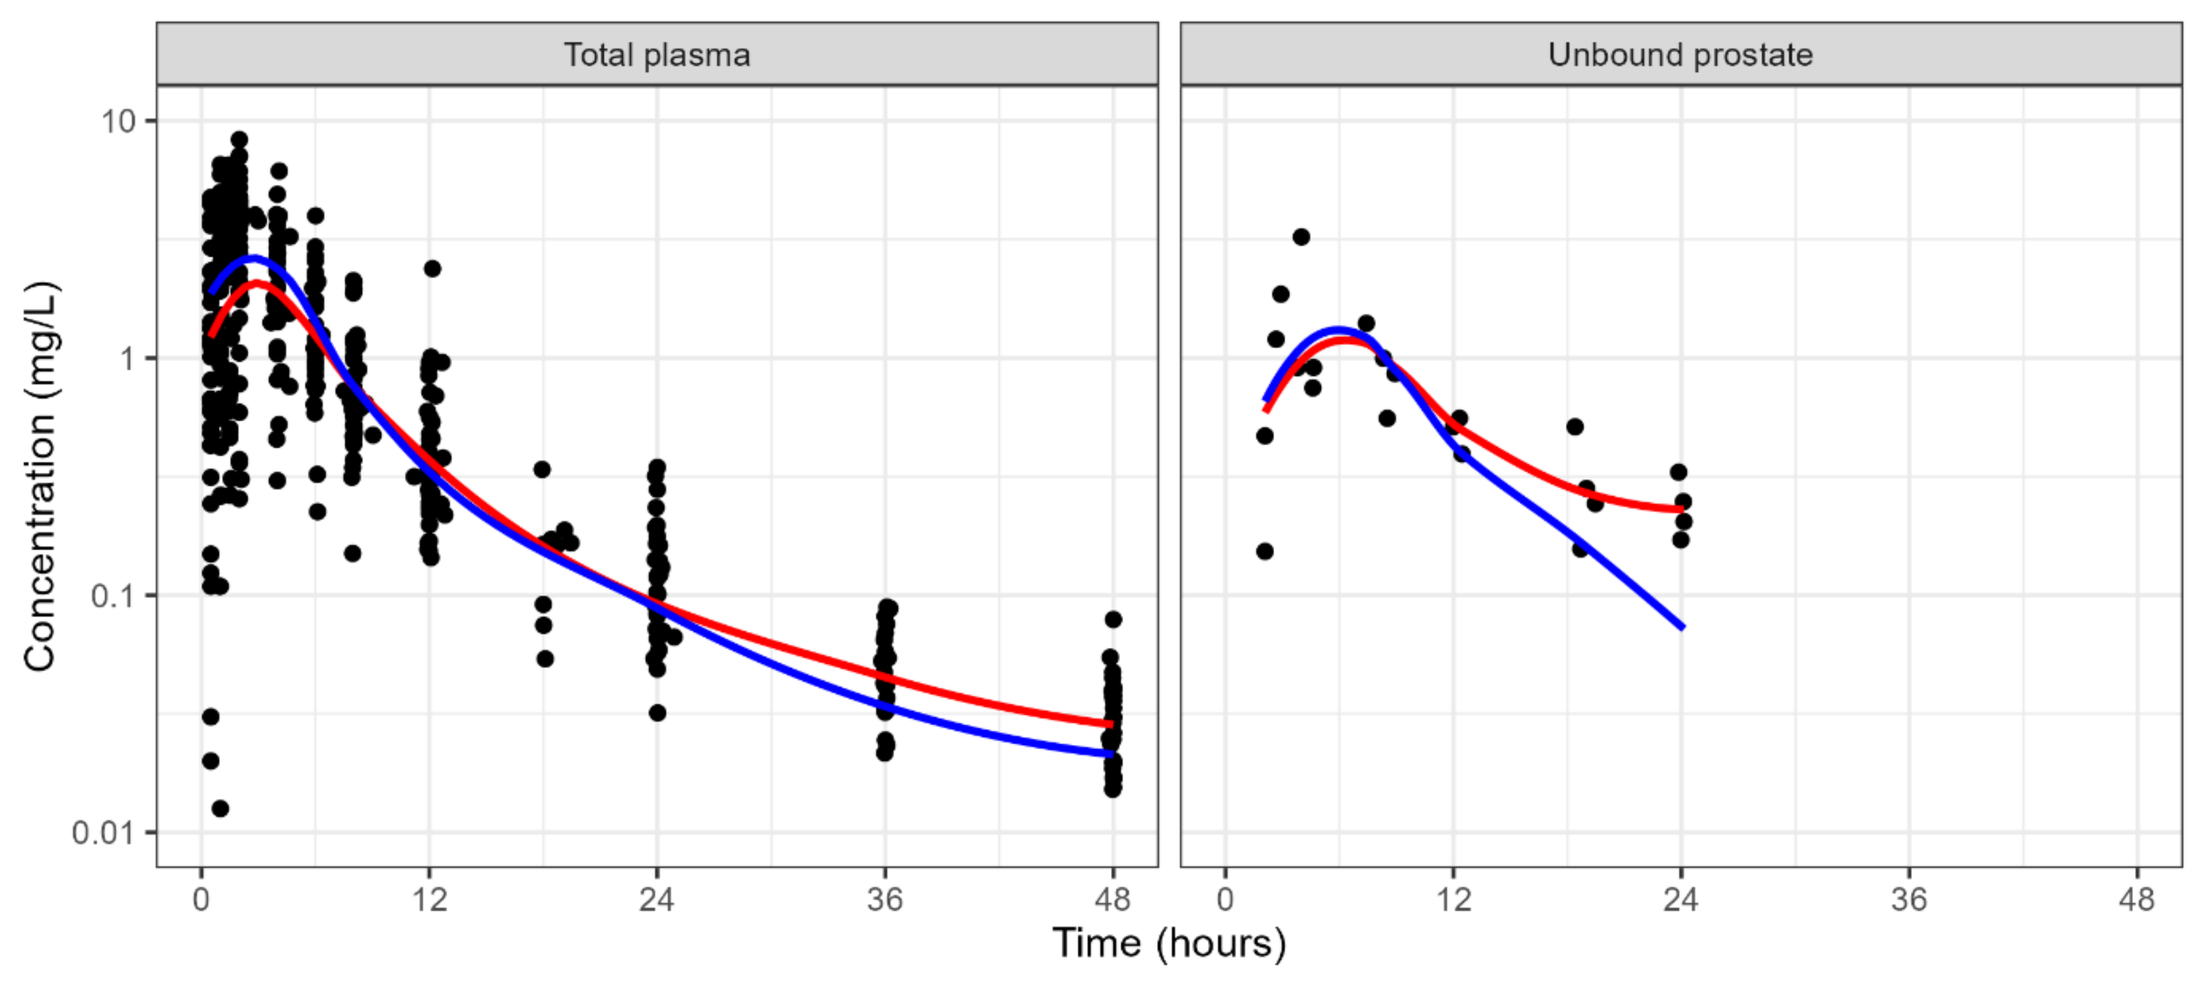
Total plasma
Unbound prostate
Concentration (mg/L)
Time (hours)

Figure 3: Human measured concentrations (points) with observation mean (red) and prediction mean (blue).

Conclusion

A PBPK model was developed with rat microdialysis data and translated to a human PBPK model to adequately predict human *ex vivo* prostate microdialysate data.

References

1) Korzekwa et al. 10.1111/cts.13314
2) Rodgers and Rowland, 10.1002/jps.20322
3) Schmitt et al. 10.1016/j.tiv.2007.09.010
4) Schmitt et al. 10.1124/dmd.120.000161
5) Monolix 2023R1, Lixoft SAS, a Simulations Plus company

Disclosure statement

GSK provided funding for the bioanalysis of samples from this study.
Hwa-Ping Feng and Karen O'Dwyer are employees of and shareholders in GSK.

This project has received funding from the Innovative Medicines Initiative 2 Joint Undertaking (JU) under grant agreement No 853976. The JU receives support from the European Union's Horizon 2020 research and innovation programme and EFPIA.

34th ECCMID EUROPEAN CONGRESS OF CLINICAL MICROBIOLOGY AND INFECTIOUS DISEASES

Barcelona, Spain
27 – 30 April 2024