Cost of care for 242 adults and children with cystic fibrosis followed up in four reference medical centers in France.

M Viprey, I Durieu, H Rouzé, S Poupon-Bourdy, Q Reynaud, S. Touzet

▶ **To cite this version:**

M Viprey, I Durieu, H Rouzé, S Poupon-Bourdy, Q Reynaud, et al.. Cost of care for 242 adults and children with cystic fibrosis followed up in four reference medical centers in France.. 40th European Cystic Fibrosis Conference, Jun 2017, Séville, Spain. hal-04332101

HAL Id: hal-04332101

https://hal.science/hal-04332101

Submitted on 8 Dec 2023

HAL is a multi-disciplinary open access archive for the deposit and dissemination of scientific research documents, whether they are published or not. The documents may come from teaching and research institutions in France or abroad, or from public or private research centers.

L'archive ouverte pluridisciplinaire **HAL**, est destinée au dépôt et à la diffusion de documents scientifiques de niveau recherche, publiés ou non, émanant des établissements d'enseignement et de recherche français ou étrangers, des laboratoires publics ou privés.

Poster Sessions / Journal of Cystic Fibrosis 16S1 (2017) S63–S174 S167

410

Costs of care for 242 adults and children with cystic fibrosis followed up in four reference medical centers in France

M. Viprey[1,2], I. Durieu[2,3], H. Rouzé[1], S. Poupon-Bourdy[1], Q. Reynaud[2,3], S. Touzet[1,2]. [1]*Hospices Civils de Lyon, Pôle IMER, Lyon, France;* [2]*Univ. Lyon, Université Claude Bernard Lyon 1, HESPER EA 7425, Lyon, France;* [3]*Hospices Civils de Lyon, Centre Hospitalier Lyon Sud, CRCM Mucoviscidose, Pierre Bénite, France*

Objectives: Costs of care are influenced by evolution of characteristics of cystic fibrosis (CF) patients due to increasing life expectancy and by recent apparition of expensive innovative therapeutics. The aim of our study was to update CF-related costs in France in 2015.

Methods: We conducted a retrospective cohort study with non-pulmonary-transplanted patients followed up in 2 adult and 2 pediatric reference medical centers in France and covered by the French National Health Insurance (NHI). Information on sociodemographic data and CF characteristics were collected from the patients' medical records. Consumptions related to CF were extracted from the regional French medical insurance health care funding database. Economic evaluation was performed from the perspective of the NHI with a time horizon of 1 year. Overall annual CF-related mean cost and annual mean cost per source of expenditure were evaluated.

Results: A total of 242 patients have given their consent for extraction of their health-care consumption data and were included. Among them 38.8% were adults and 46.3% were females. Median age was 15.0 (min: 0; max: 65). Children were distributed in 21.1% aged 0–5 years old, 21.9% aged 6–11 years old and 18.2% aged 12–17 years old. The mean overall annual CF-related cost per patient was €27 315. Hospital care represents 30.5% of costs and home care 69.5% of costs, among them 46.2% for drugs and 8.4% for physiotherapists' consultations. Concerning drug consumption, 93.8% of patients had at last 1 dispensing of oral or intravenous antibiotic, 38.0 of inhaled tobramycin, 26.9% of inhaled colymicin, 54.1% of dornase alpha, 83.9% of pancreatic enzymes. Four patients were on ivacaftor with a mean overall annual CF-related cost per patient of €204 395.

Conclusion: CF-related costs are increasing since 2003 where a mean overall annual CF-related cost per patient of €22 725 was evaluated. Major sources of expenditure stay drugs consumptions and hospital care.

411

Disease progression in patients (pts) with cystic fibrosis (CF) treated with ivacaftor (IVA): analysis of real-world data from the UK CF Registry

L. Bessonova[1], N. Volkova[1], M. Higgins[2], L. Bengtsson[1], S. Tian[1], C. Simard[1], S.O. Nyangoma[3], D. Bilton[3,4]. [1]*Vertex Pharmaceuticals Incorporated, Boston, United States;* [2]*Vertex Pharmaceuticals (Europe) Limited, London, United Kingdom;* [3]*Imperial College London, London, United Kingdom;* [4]*UK CF Registry, London, United Kingdom*

Objectives: This ongoing, long-term, postapproval observational safety study uses existing CF registries' data to evaluate outcomes in pts treated with IVA in a real-world setting. Here we present results of disease progression analyses using 2015 data from the UK CF Registry.

Methods: Pts with a record of IVA use in the first year of commercial availability in the UK (2013), who continued treatment through 2015 and had no organ transplantation, were compared to comparator (COMP) pts with no IVA use record matched (≈1:5) on age, sex, and genotype severity (eg, *G551D* mutation IVA pts were primarily matched to pts homozygous for *F508del* ineligible for IVA therapy). Outcomes included percent predicted FEV_1 ($ppFEV_1$), pulmonary exacerbations (PEx), PEx hospitalizations, body mass index (BMI), distal intestinal obstruction syndrome (DIOS), and CF-related diabetes (CFRD).

Results: IVA (N = 260) and COMP (N = 1,289) pts were similar in age, sex, lung function, and medication use in 2012. From 2012 to 2015, mean $ppFEV_1$ change was +5.8 percentage points in IVA vs –3.1 percentage points in COMP pts; mean BMI increase was 1.7 kg/m^2 for IVA vs 0.6 kg/m^2 for COMP pts. The proportion of IVA pts experiencing PEx decreased from 54.6% to 35.4%, compared to an increase from 46.3% to 56.4% in COMP pts. Other outcomes are summarized in the Table.

Conclusion: In addition to maintained improvements in lung function and nutritional status, as well as decreased risk of PEx and PEx hospitalizations, pts treated with IVA had a lower prevalence of DIOS and CFRD relative to COMP pts. These results support disease modification by CFTR modulator therapy.

Acknowledgement: Sponsored by Vertex Pharmaceuticals Incorporated

Table:

	$ppFEV_1$, mean (SD)		Risk of PEx Hospitalizations, %	DIOS Prevalence, %	CFRD Prevalence, %	
	IVA	COMP	IVA \| COMP	IVA \| COMP	IVA \| COMP	IVA \| COMP
2012, baseline	71.9 (24.2)	72.5 (22.9)	54.6 \| 46.3	47.7 \| 42.3	6.5 \| 7.1	16.9 \| 22.0
2013	78.6 (25.2)	72.9 (23.3)	48.1 \| 56.0	36.9 \| 43.0	4.6 \| 7.1	18.1 \| 24.5
2014	79.1 (25.0)	71.7 (23.5)	32.3 \| 56.3	22.7 \| 44.4	3.9 \| 7.7	20.0 \| 27.3
2015	78.1 (25.6)	70.5 (24.2)	35.4 \| 56.4	28.1 \| 44.6	3.1 \| 7.7	21.2 \| 30.6

412

Spanish multicentre registry-based study on the phenotype of patients with cystic fibrosis carrying the V232D (c.695T>A) mutation

M.D. Pastor[1], C. Vazquez[2], M.E. Quintana-Gallego[3], I. Delgado[4], L. Máiz Carro[5], V. Sanz[6], A. Aguilar[7], R.M. Girón-Moreno[8], C. Luna[9], J.R. Villa[10], C. Prados[11], M.I. Barrio[12], S. Gartner[13], O. Asensio[14], A. Escribano[15], M. Cols[16], A. Gómez[17], J.I. Pijoan[18]. [1]*Hospital de Cruces, Pediatrics, Bizkaia, Spain;* [2]*Hospital de Cruces, Paediatrics, Bizkaia, Spain;* [3]*Hospital Virgen del Rocío, Pulmonology, Sevilla, Spain;* [4]*Hospital Virgen del Rocío, Paediatrics, Sevilla, Spain;* [5]*Hospital Ramón y Cajal, Pulmonology, Madrid, Spain;* [6]*Hospital Ramón y Cajal, Paediatrics, Madrid, Spain;* [7]*Hospita Infantill Las Palmas, Paediatrics, Las Palmas, Spain;* [8]*Hospital La Princesa, Pulmonology, Madrid, Spain;* [9]*Hospital 12 de Octubre, Paediatrics, Madrid, Spain;* [10]*Hospital Infantil Niño Jesús, Paediatrics, Madrid, Spain;* [11]*Hospital La Paz, Pulmonology, Madrid, Spain;* [12]*Hospital La Paz, Paediatrics, Madrid, Spain;* [13]*Hospital Vall d¨Hebrón, Paediatrics, Barcelona, Spain;* [14]*Hospital Parc Taulí, Paediatrics, Sabadell, Spain;* [15]*Hospital Clinic, Paediatrics, Valencia, Spain;* [16]*Hospital Sant Joan de Déu, Paediatrics, Barcelona, Spain;* [17]*Hospital de Cruces, Pulmonology, Bizkaia, Spain;* [18]*Hospital de Cruces, Epidemiology, Bizkaia, Spain*

Objectives: To investigate the clinical impact (so far unknown) of V232D, a valine to aspartate change in exon 6a reported in 1993 [1].

Methods: Out of the 2015 national dataset of 2003 patients with CF under care at 19 CF centres participating in the ECFSPR, we identified 29 patients carrying the V232D allele and: F508del (14), other CF-causing mutations (12), or another unknown mutation (3). Clinical variables in patients with V232D (V232Dp), and in all 1718 Spanish patients on the 2014 Spanish national dataset (allCF) were compared

Results: V232D accounted for 0.72% of Spanish CF alleles. V232Dp were older at diagnosis 9.2 ± 15.3 years (median 1.1) vs 4.5 ± 9.9 (median 1.1), $p = 0.048$. Age at follow-up was not different (20.6 ± 16 vs 18.6 ± 13). Just 4 V232Dp had been diagnosed by newborn screening. Sweat chloride (mmol/l) was lower: 71.7 ± 17.2, range 40–102, vs 90.4 ± 26 ($p < 0.001$). There was no difference in the proportion of patients with sweat chloride ≥60 and < 60 mmol/L. One single V232Dp was on pancreatic enzymes vs 1105 (73%), p < 001. FEV_1%Pred. was 87.9 ± 20.2 in V232Dp vs 79.3 ± 24.7 in allCF ($p = 0.098$). It was not different in patients aged < 18 (93.4 ± 16 vs 92.6 ± 20, but adult V232Dp fared better (83.6 ± 22 vs 67.4 ± 22 ($p = 0.01$). No V232Dp younger than 18 had chronic bronchial infection with *P aeruginosa* (*cPa*) vs 121(13.8%) in allCF (NS), while 3/13 (23.1%) adults had *cPa* vs 626/1478 (42.4%), ($p = 0.01$). There was no difference between groups in the number of patients with chronic bronchial infection with *S aureus*.

Conclusions: Most V232D patients had classical CF albeit with comparatively lower sweat chloride values. They were nearly always PS, and their average age at diagnosis was higher. Adults had significantly better FEV_1 and lower prevalence of *cPa*. Still 6/2 (26%) of those with available pulmonary function data, had FEV_1% pred 50–69%, so close follow-up is warranted as a sizeable minority may develop substantial pulmonary disease.

Reference

1. www.genet.sickkids.on.ca